POST-HOC ANALYSIS OF THE 13-VALENT POLYSACCHARIDE CONJUGATE VACCINE EFFICACY AGAINST VACCINE-SEROTYPE PNEUMOCOCCAL COMMUNITY ACQUIRED PNEUMONIA IN AT-RISK OLDER ADULTS

Authors: Suaya JA[1], Jiang Q[1], Bonten M[2], Patterson S[3], van Werkhoven H[2], Scott D[4], Gruber W[4], Webber C[5], Schmoele-Thoma B[6], Hall-Murray C[1], Sylvester G[1], Jodar L[1], Isturiz R[1]

[1]Pfizer Vaccines Medicines Development & Scientific and Clinical Affairs, Collegeville, PA, U.S.; [2]University Medical Centre Utrecht, Utrecht, The Netherlands;
[3]Pfizer Vaccines Clinical Research & Development, Collegeville, PA, U.S.; [4]Pfizer Vaccine Clinical Research, Pearl River, NY, U.S.;
[5]Pfizer Vaccine Clinical Research, Walton Oaks, U.K.; [6]Pfizer Vaccine Clinical Research, Berlin, Germany;

Presented at ISPPD, 2016. 10th International Symposium on Pneumococci and Pneumococcal Diseases. Poster Session #: *PO06a "The Unique Challenge of Pneumococcal Disease in Adults, Pneumococcal Disease in Adults*." Poster #634. Date: June 29, 2016 . Glasgow, United Kingdom

Background and Aim

- The Community Acquired Pneumonia Immunization Trial in Adults (CAPiTA) was a randomized, double-blind, placebo-controlled clinical trial with 84,496 subjects randomized.
- CAPiTA demonstrated that vaccination of adults ≥65 years of age with 13-valent pneumococcal polysaccharide conjugate vaccine (PCV13) was efficacious in preventing vaccine-type (VT) *Streptococcus pneumoniae* community-acquired pneumonia (VT-CAP) and VT invasive pneumococcal disease.[1]
- Public health authorities such as the U.S. Advisory Committee on Immunization Practices have been interested in knowing the performance of PCV13 in populations known to have higher risk of CAP (at-risk).
- The current study is a post-hoc exploratory analysis of PCV13 vaccine efficacy (VE) performed by stratifying participants in CAPiTA by their at-risk status.

Methods

- All participants were asked to complete a questionnaire with a screening list of medical conditions before they received the study vaccine: heart disease, lung disease, asthma, diabetes with insulin use, diabetes without insulin use, liver disease, smoking, and history of spleen removal.
- Self-identified medical conditions were not verified by medical record review.
- Subjects were excluded from this analysis if they self-identified with a history of splenectomy or if participants did not respond to any of the questions in the screening list of the medical conditions form.
- "At-risk" classification was based on at least one of the other self-identified medical conditions.
- Participants without any at-risk condition were classified as "without known-risk."
- Outcomes of interest included PCV13 VE against VT-CAP and against *Streptococcus pneumoniae* CAP (Sp-CAP) in at-risk and without-known-risk groups.
- The same case definitions (for VT-CAP, Sp-CAP), analyses (per-protocol of first episode), and statistical methods (calculation for VE and confidence interval [CI]) were applied as in the original CAPiTA publication.[1]

Results

- Of the 84,496 participants in the original efficacy analysis, 75 were excluded from the post-hoc analysis. (Table 1)
- Of those included in the current analysis, 41,590 were at-risk and 42,831 were without-known risk . Each group was distributed evenly between PCV13 and placebo groups. (Table 1)
- The prevalence of each self-identified medical condition was: heart disease, 25.4% ; diabetes, 12.5%; smoking, 12.3% ; lung disease, 10.2%; asthma, 4.9%; liver disease, 0.5%, with similar prevalence in PCV13 and placebo groups.
- VT-CAP was identified in 115 at-risk participants (43 and 72 in the PCV13 and placebo groups, respectively). PCV13 VE: 40.3% (95.2% CI: 11.4%, 60.2%). (Table 3)
- Sp-CAP was identified in 197 at-risk participants (81 and 116 in the PCV13 and placebo groups, respectively). PCV13 VE: 30.2% (95%CI: 6.5%, 48.1%). (Table 3)
- VT-CAP was identified in 24 participants without known-risk (6 and 18 in the PCV13 and placebo groups, respectively). PCV13 VE: 66.7%; (95.2% CI: 11.7%, 89.3%). (Table 3)
- Sp-CAP was identified in 47 participants without known-risk (19 and 28 in the PCV13 and placebo groups, respectively). PCV13 VE: 32.1% (95%CI:-25.9%,64.2%). (Table 3)

Results (cont.)

Table 1. Population Attrition and At-Risk Classification in Post-hoc Analysis of PCV13 Vaccine Efficacy in Adults

	PCV13		Placebo		Total	
	Participants	%	Participants	%	Participants	%
Total participants in the original efficacy analysis[1]	42,240		42,256		84,496	
Exclusions from this post-hoc analysis						
History of splenectomy	41	0.1%	32	0.1%	73	0.1%
Incomplete medical condition form	1	0.0%	1	0.0%	2	0.0%
Included in the post-hoc analysis	42,198	99.9%	42,223	99.9%	84,421	99.9%
At-risk classification						
Without-known risk	21,418	50.8%	21,413	50.7%	42,831	50.7%
At-risk participants	20,780	49.2%	20,810	49.3%	41,590	49.3%

Note: Percentages (%) for exclusions are as % of total participants; for at-risk classification are as % of participants included in the post-hoc analysis.

Table 2. Baseline Characteristics of Participants in Post-hoc Analysis of PCV13 Vaccine Efficacy in Adults

	At Risk (N= 41,590)				Without Known-Risk (N= 42,831)			
	PCV13 (N= 20,780)		Placebo (N= 20,810)		PCV13 (N= 21,418)		Placebo (N= 21,413)	
	Participants	%	Participants	%	Participants	%	Participants	%
Sex								
Female	8,344	40.2%	8,125	39.0%	10,431	48.7%	10,318	48.2%
Male	12,436	59.8%	12,685	61.0%	10,987	51.3%	11,095	51.8%
Race								
White	20,435	98.3%	20,470	98.4%	21,126	98.6%	21,113	98.6%
Non-White	341	1.6%	333	1.6%	287	1.3%	297	1.4%
Missing	4	0.0%	7	0.0%	5	0.0%	3	0.0%
Age at enrollment								
Mean (SD)	73.1 (5.8)		73.1 (5.8)		72.5 (5.6)		72.4 (5.5)	
Median	72.0		72.0		71.2		71.1	
Min - Max	64.9 - 101.1		64.9 - 99.5		61.9 - 100.3		63.3 - 96.1	
< 75 Years	13,729	66.1%	13,720	65.9%	15,254	71.2%	15,322	71.6%
75-<85 Years	6,240	30.0%	6,311	30.3%	5,474	25.6%	5,433	25.4%
≥85 Years	811	3.9%	779	3.7%	690	3.2%	658	3.1%

Table 3. Post-hoc Analysis of PCV13 Vaccine Efficacy in Adults

	Participants	PCV13 VT-CAP		*sp-* CAP	
		Estimate	95.2% CI	Estimate	95.0% CI
All partipants[1]	84,496	45.6%	(21.8% , 62.5%)	30.6%	(9.8% , 46.7%)
At-risk classification					
At-risk	41,590	40.3%	(11.4% , 60.2%)	30.2%	(6.5% , 48.1%)
Without-known risk	42,831	66.7%	(11.7% , 89.3%)	32.1%	(-25.9% , 64.2%)

Abbreviations: PCV13 vaccine-type (VT) streptococcus pneumoniae community-acquired pneumonia (VT-CAP); *streptococcus pneumoniae* community-acquired pneumonia (*sp* -CAP); CI, confidence interval.

Discussion

- In this post-hoc exploratory analysis, there were statistically significant estimates of PCV13 efficacy against VT-CAP and Sp-CAP in participants at-risk, that were overall similar to those estimates for the entire study participants.[1]
- In participants without-known risk, the point estimate of PCV13 efficacy against VT-CAP was higher than in participants at-risk: PCV13 VE: 67.7%; (95.2% CI: 11.7%, 89.3%) vs. 40.3% (95.2% CI: 11.4%, 60.2%).
- PCV13 efficacy against Sp-CAP in participants at-risk was similar to participants without-known risk. However, no statistical significance was observed among the participants without-known risk.

Limitations

- Because of the nature of post-hoc analysis (unplanned up front), results should be seen as exploratory and hypothesis generating.
- Medical conditions were self-reported and lacked medical record verification.
- Medical conditions were not mutually exclusive.
- Study was not designed or powered to estimate VE in sub-populations.

Conflicts of interest:
All authors except MB and HvW were Pfizer employees at the time of this post-hoc analysis and poster development.

References:
1. Bonten et al. N Engl J Med 2015:372;12