Phoenix Nest: Starting A Small Company Focused on Sanfilippo Syndrome

Jill Wood[1,2] and Sean Ekins[1]

[1]Phoenix Nest, P.O. BOX 150057, Brooklyn NY 11215, USA., [2]Jonah's Just Begun, P.O. Box 150057, Brooklyn, NY 11215, USA.

Abstract

Sanfilippo syndrome (mucopolysaccharidosis type III; MPS III) is a devastating neurodegenerative lysosomal storage disorder of childhood. There is currently no cure or effective treatment available for MPS III. There are however many therapies in early development, including gene therapies, enzyme replacement, chaperone and substrate reduction. With Sanfilippo MPSIII Type A, there is currently a large focus on gene therapy evaluating different vectors across many different groups. Less research appears to be focused on types C and D. Phoenix Nest (PN) was formed by a parent (of a child with MPSIIIC) and a pharmaceutical researcher. Initially the goal was to create a virtual company that would then be able to apply for STTR and SBIR grants to work on treatments for MPS types C and D. In the space of 2 years we have built up collaborations with leading academics and industry and submitted multiple grant proposals. We were able to secure our first STTR recently and this sets the stage for our growth. All of our resources are spent on supporting research and development with minimal overhead as we leverage collaborative researchers and tools to ensure we reach our goal. We will describe our strategy and propose that alongside forming foundations, a patient driven company may be a useful vehicle to push for more translational research in a rare disease. We would encourage other parents and researchers to start a small company and learn from this experience. Due to the limited pool of funding for this disorder, enhanced collaboration between foundations, academics and companies facilitated by small companies like PN may prevent unnecessary redundancies and broaden the impact of the ongoing research efforts. Ultimately our goal is to deliver a treatment to the patient. We will summarize the current state of research for Sanfilippo syndrome and describe the path we have taken and the pros and cons of forming a company along with some recommendations. As we are just beginning PN we see our own collaboration as an experiment that if successful could be a useful model for other rare diseases.

Introduction

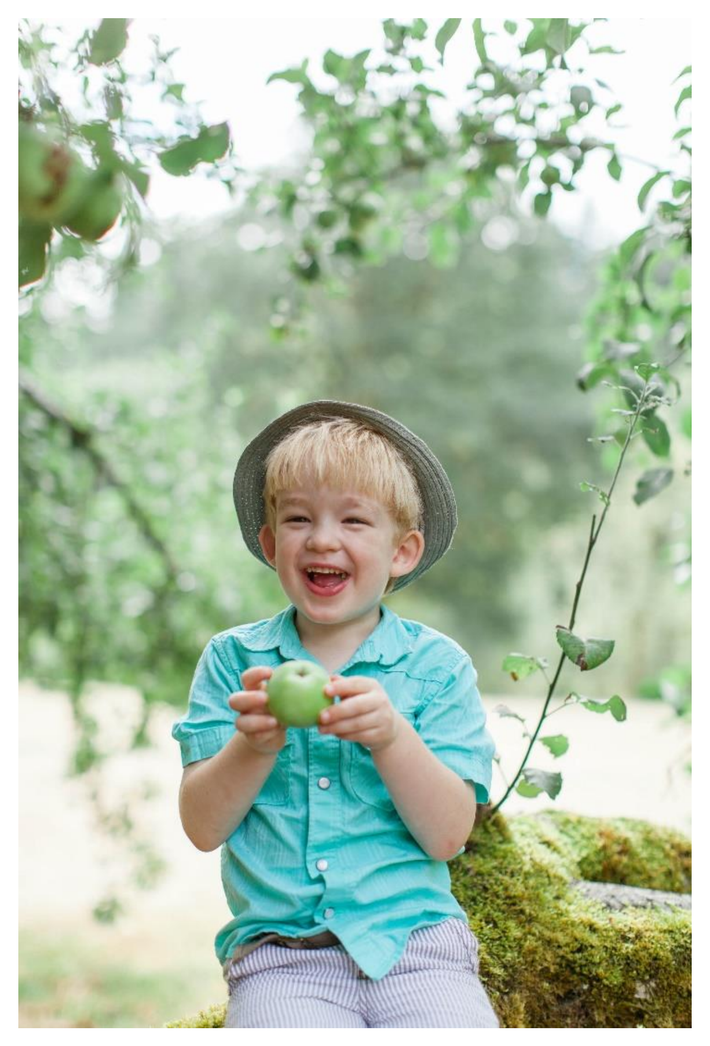

Jonah (6), diagnosed age 2.

Has Sanfilippo Syndrome (MPS IIIC) - MPS IIIC caused by genetic deficiency of heparan sulfate acetyl CoA: α-glucosaminide N-acetyltransferase, (HGSNAT). **A single gene defect.**

MPS III is a devastating neurodegenerative lysosomal storage disorder of childhood.

All subtypes of MPS III have similar clinical phenotypes with onset in infancy or early childhood: progressive and severe neurological deterioration, hearing loss, and visceral manifestations. Most patients become demented and die before adulthood but some survive to the fourth decade with progressive dementia and retinitis pigmentosa.

Jonah's parents founded Jonah's Just Begun 501(c)3

Funds raised to invest in the science of MPS IIIC.

It took at least 1 year to understand the state of the science and build connections with scientists.

From a modest start - now part of the global HANDS consortium.

Sanfilippo foundations fund global research…

Now funding multiple treatments, including gene therapy…

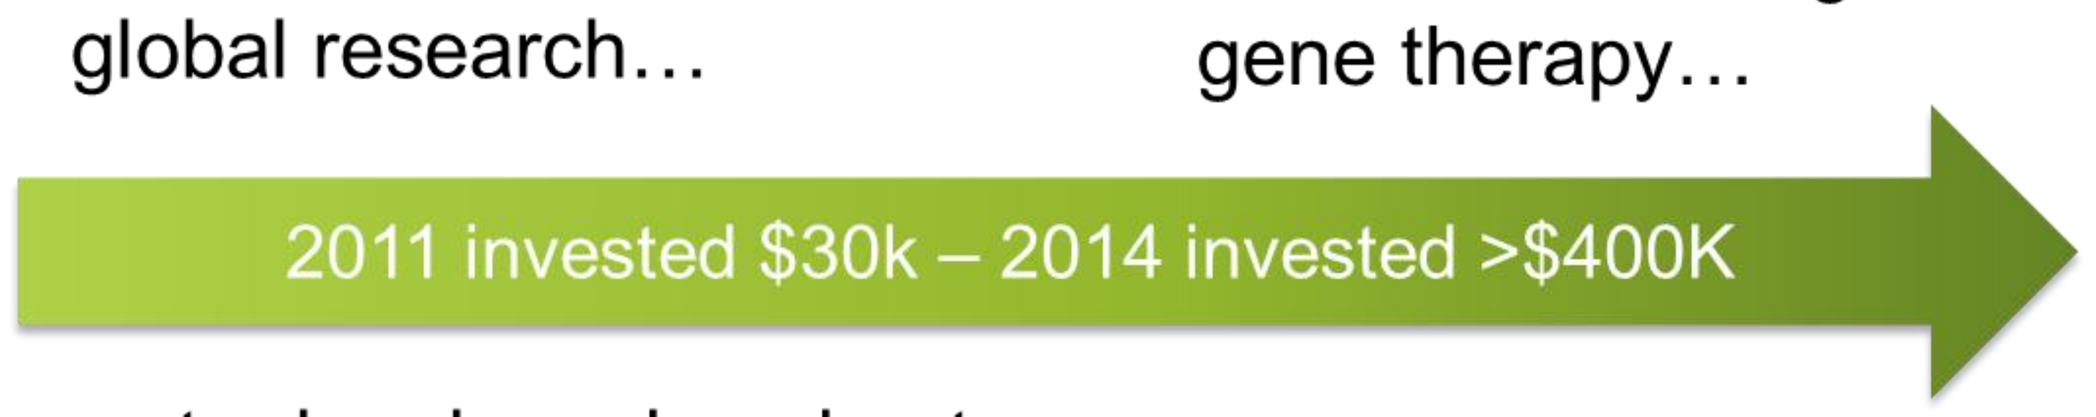
2011 invested $30k – 2014 invested >$400K

…but treatment is still years away

…to develop a knockout mouse that models human Sanfilippo in an animal model.

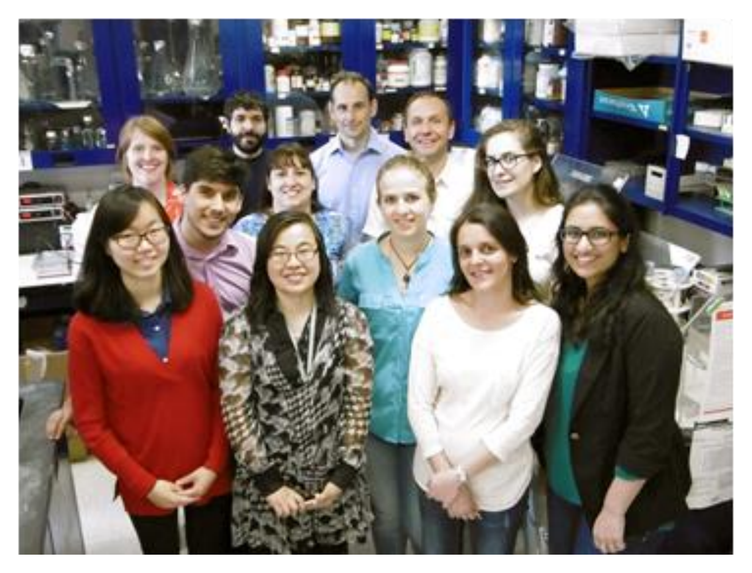

Other rare diseases are not so lucky

How do we scale and accelerate progress to 1000's of understudied diseases?

Rare Disease Parent Odyssey

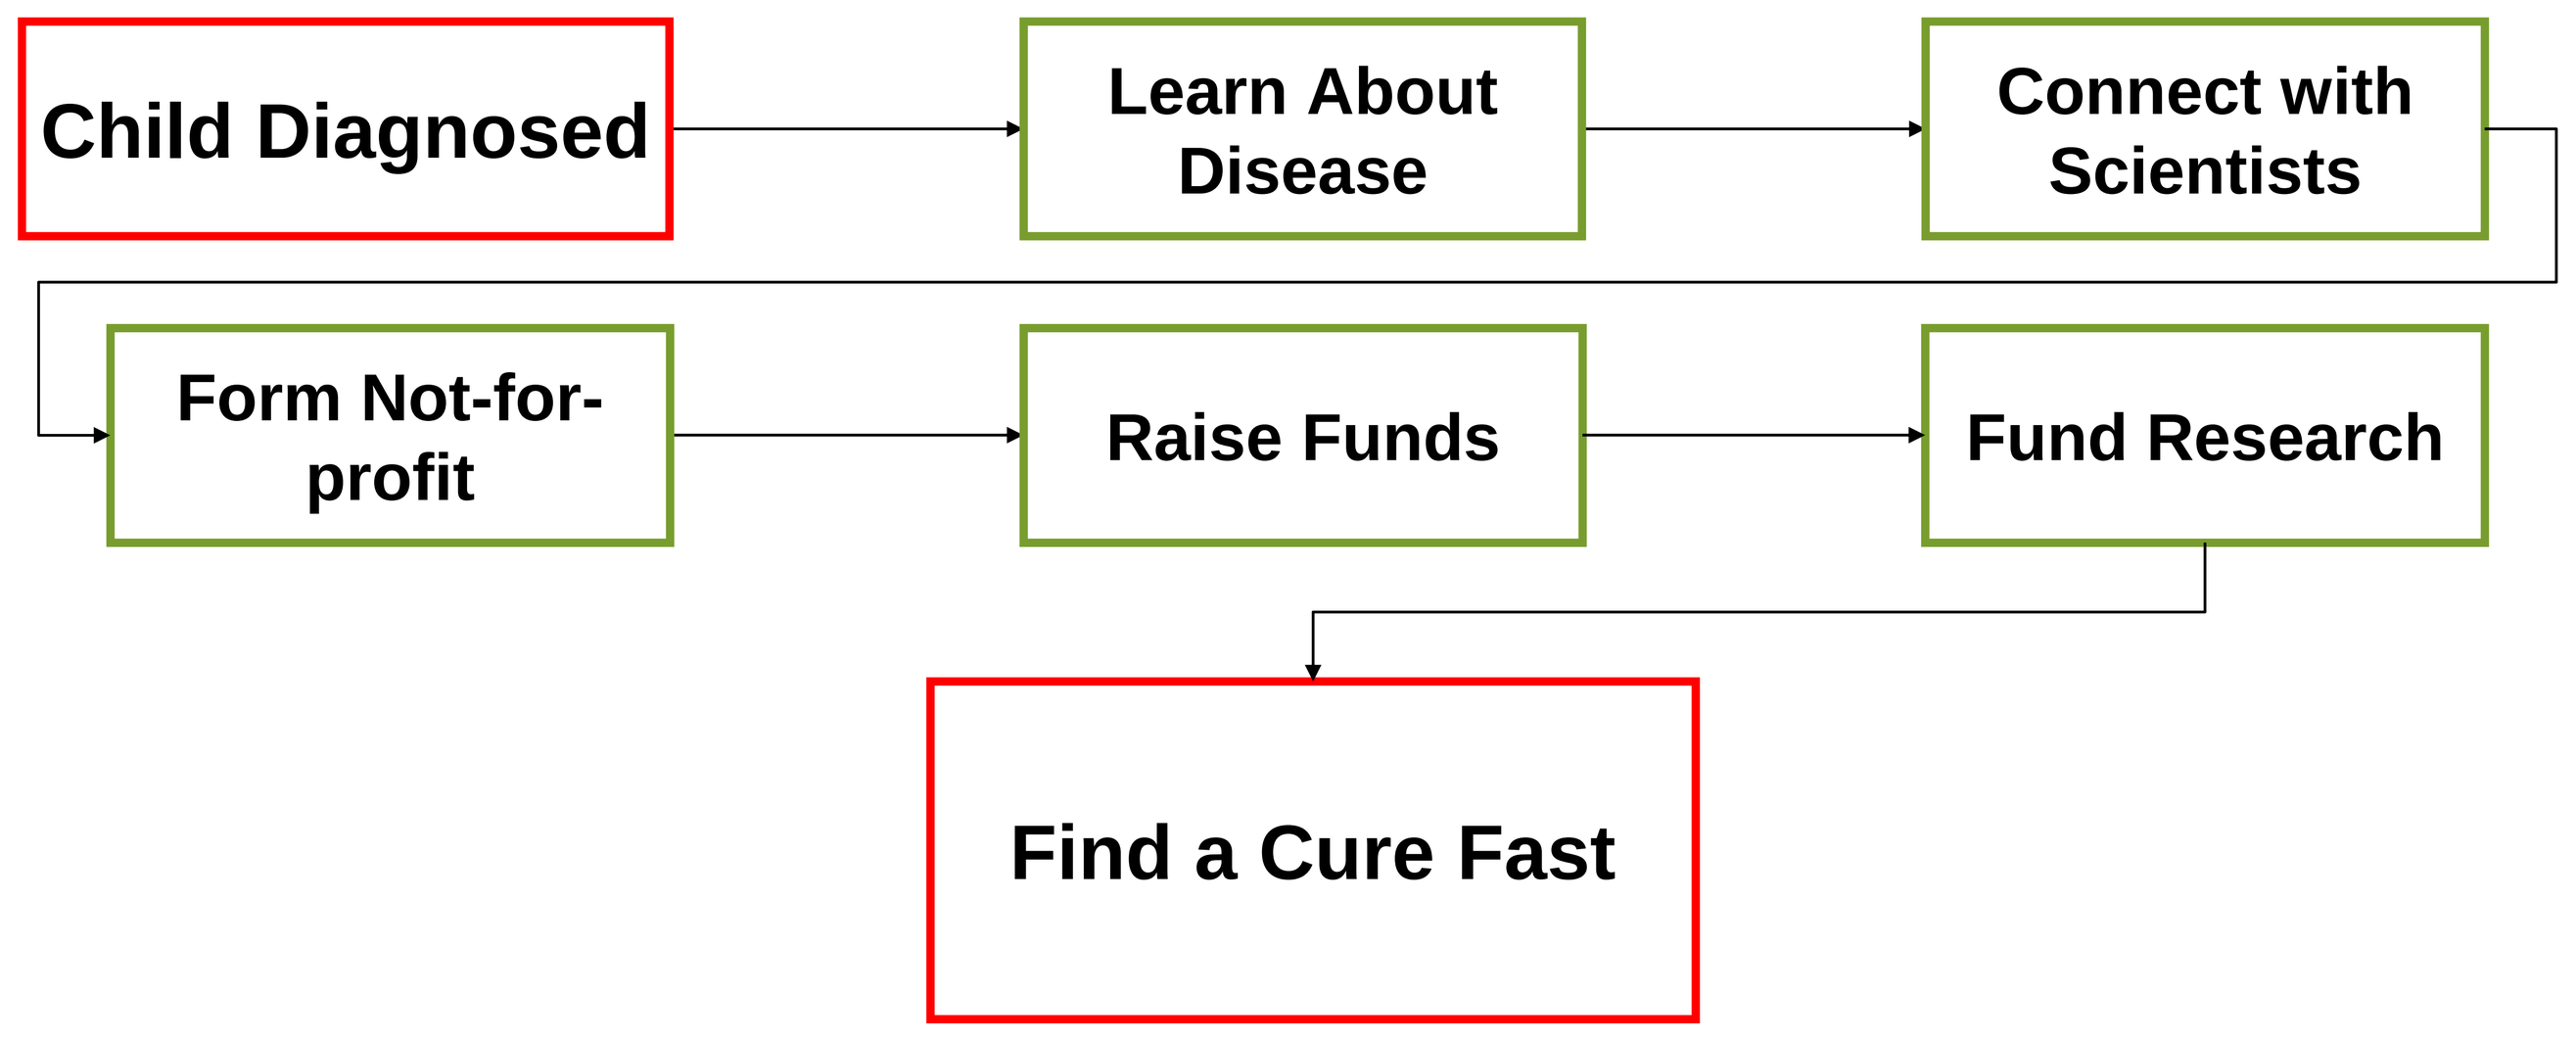
Child Diagnosed
Learn About Disease
Connect with Scientists
Form Not-for-profit
Raise Funds
Fund Research
Find a Cure Fast

Timeline: Starting a Biotech Company

Nov 2011 – JW and SE met at Partnering for Cures – SE suggested a company would provide a means to go after SBIR and STTR grants to fund research.

Early 2012 – Phoenix Nest (PN) founded by Jonah's parents.

April 2012 – PN submitted first grant with collaborator.

Dec 2012 – PN resubmitted first grant (ultimately not funded) and JJB submitted proposals for Rare Genomics Institute and Assay Depot Rare Disease Science Challenge: Be HEARD Competition.

Feb 2013 – Funded Sanfilippo D knock out mouse study (a prize from Rare Disease Science Challenge: Be HEARD).

Nov 2013 – Published paper with other rare disease advocates.

Dec 2013 –Submitted a second new STTR with collaborator, also submitted third STTR with Patti Dickson (LABioMed) on ERT.

Aug 2014 – Submitted a fourth and fifth STTR.

Oct 2014 – STTR with Patti Dickson funded and started.

Dec 2014 - One grant scored and the other triaged.

Challenges of Starting a Biotech Company

Starting a company needs good legal and accounting advice, but is relatively easy to do.

JW was able to make the transition from skills picked up at former retail jobs. e.g. record keeping, Quickbooks, self audits, PO reconciliation and the HR side (new hire paperwork, payroll, health insurance). In retail management you hire, fire, maintain EOM, self audits, take in inventory, work with customers and vendors). JJB taught JW the board of directors process (minutes, budgets, bank accounts, fundraising etc).

We are funding company by obtaining STTR and SBIR grants.

Registering company on multiple websites in order to submit grants:
- Dun and Bradstreet Universal Numbering System
- System for Award Management
- SBA Company Registry
- eRA Commons
- Grants.gov

This is time consuming and appears excessive - why not consolidate?

Company needs a qualified scientist (PI) who can coordinate grant writing with academic and submit grant package for company. In addition must have credibility at managing science.

Reviewers of rare disease grants do not understand that many key scientists are outside the US and their decision can prevent collaboration and progress.

Reviewers' attitude towards parents submitting grants needs improvement even though NIH encourages us to take this route.

Pipeline for Sanfilippo Syndrome Therapeutics

Sanfilippo Syndrome Type	Treatment	Company Group
A	Gene Therapy SAF-301 completed phase I/II	Lysogene
A	Gene therapy	Nationwide Children's Hospital / Abeona Therapeutics
A	Gene therapy	Esteve
A	Enzyme replacement therapy	Shire
A	Stem cells	Univ. Manchester
A	Substrate reduction therapy Genistein	Univ. Manchester
B	Gene therapy ongoing phase I/II	Pasteur Inst.
B	Gene therapy	Nationwide Children's Hospital / Abeona Therapeutics
B	Enzyme replacement therapy	Biomarin / Synageva
B	Substrate reduction therapy Genistein	Univ. Manchester
C	Chaperone	Univ. Montreal
C	Gene Therapy	Univ. Manchester
D	**Enzyme replacement therapy**	**LaBioMed / Phoenix Nest**

Our Strategy

When our STTR grant was funded we had further steps to take to comply with NIH rules. This required considerable effort.

Since obtaining a grant we have had extensive legal contract work to license technology and set up a laboratory close to collaborator.

Hiring a postdoc was tougher than anticipated with 2 rejections.

Setting up contracts is a major undertaking.

Working on Sanfilippo Syndrome Type D should help Type C research.

Synergy with Knock out mouse work being undertaken.

In addition we are licensing early stage therapeutics for Type C.

The Future

Next step will be submitting a phase II STTR grant in 2015. This will be a huge undertaking. It will ultimately lead to producing clinical scale levels of protein.

We will need skill sets outside of those currently in the company.
If successful this could grow the company.
Ultimately may need to partner to fund clinical trials.

Other licensed therapeutics should give company a broad platform for MPS III.

Beyond Phoenix Nest

Apply our strategy to multiple rare diseases.

Challenge will be identifying technology that could benefit from STTR / SBIR grants. How do we connect scientists and parents-entrepreneurs?

Raise awareness that forming a company is a valid approach and can help scientists fund their research and translate to the clinic.

Acknowledgments

Dr. Alexey Pshezhetsky, Dr. Patti Dickson, Dr. Xiaoyi Zhang, Dr. Tsui-Fen Chou, Dr. Brian Bigger., Dr. Matt Ellinwood and many others.

References

Wood J, Sames L, Moore A, Ekins S, The multifaceted roles of rare disease parent / patient advocates in drug discovery, Drug Disc Today, 18: 1043–1051, 2013. http://www.drugdiscoverytoday.com/download/1215

Jonah's Just Begun
www.jonahsjustbegun.org

www.phoenixnestbiotech.com
Phoenix Nest INC
Finding treatments for families.